Novel, Non-Addictive Opioid Dimer DMX-101 Targets Peripheral Opioid Receptors For Treatment of Chronic Pain without Risks Associated with Traditional Opioids

Authors: Nikhilesh Singh, PhD; Frank J. Steinberg, DO; Virginia Sanders, PhD

Affiliation: DIMERx, Inc.

Background

Although traditional opioids are effective in treating chronic severe pain, they are associated with significant CNS-mediated side effects and exposure of patients to risk of dependence [1]. Exclusive targeting of peripheral opioid receptors without CNS exposure should provide safe and effective management of chronic pain without the risk of addiction [2,3]. Despite up to 80% of opioid receptors located outside the CNS also mediating opiate analgesic effects [4], there is no available drug that acts at peripheral opioid receptor sites for the management of chronic pain.

DMX-101 is a new chemical entity created by covalently linking two molecules of buprenorphine. After oral administration, DMX-101 is rapidly absorbed and distributed, but its high molecular weight prevents its crossing the blood-brain barrier, avoiding the unwanted effects of CNS involvement. **Availability of a non-addictive opioid analgesic will also prevent initiation of traditional opioid use, a potential gateway to addiction.**

The following studies demonstrate DMX-101's efficacy in treating acute and neuropathic pain as well as safety and efficacy in the clinic.

Paradigm: **Mustard oil-induced inflammatory colorectal tissue damage in mice**
Treatments: Day 1 single dose, oral gavage DMX-101 40 mg/kg and placebo
Day 2 and 7 all cohorts receive placebo
Assessment: Pain behavior measured by increased abdominal licking and withdrawal
Time: One hour post-treatment on Days 1, 2, 7
Results: DMX-101 controlled pain due to hypersensitivity and hyperalgesia significantly better than placebo

Fig. 1

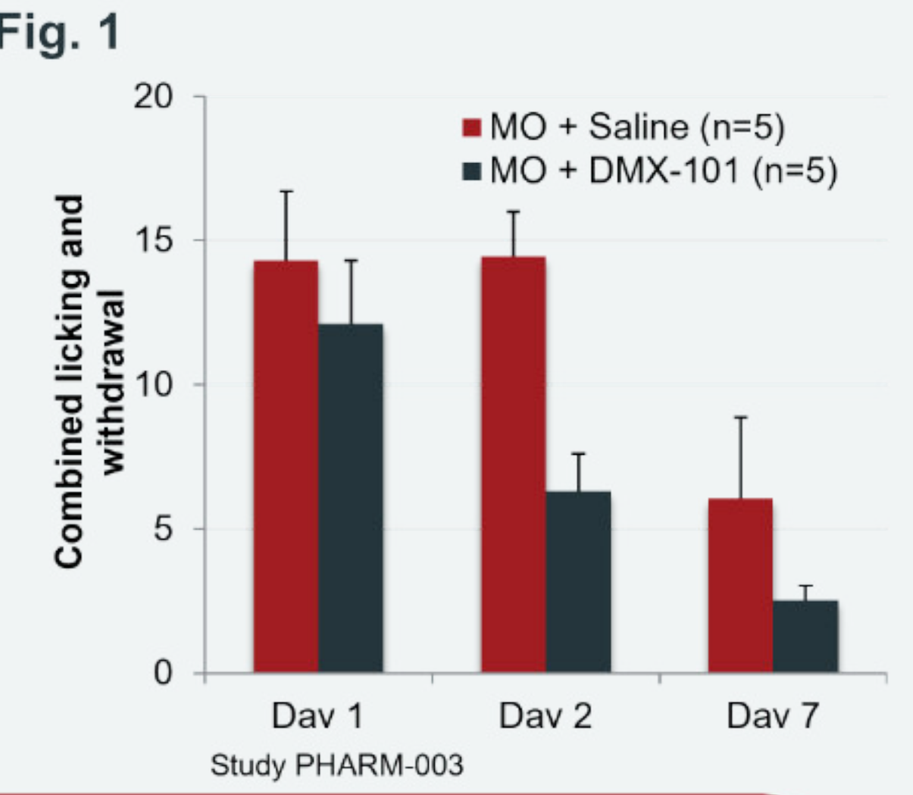
MO + Saline (n=5)
MO + DMX-101 (n=5)
Combined licking and withdrawal
Day 1
Day 2
Day 7
Study PHARM-003

Projected Clinical Benefit: Relief from abdominal pain following intestinal tissue damage

Study 1: Hyperalgesic Effect of DMX-101 vs. Placebo

Fig. 2a

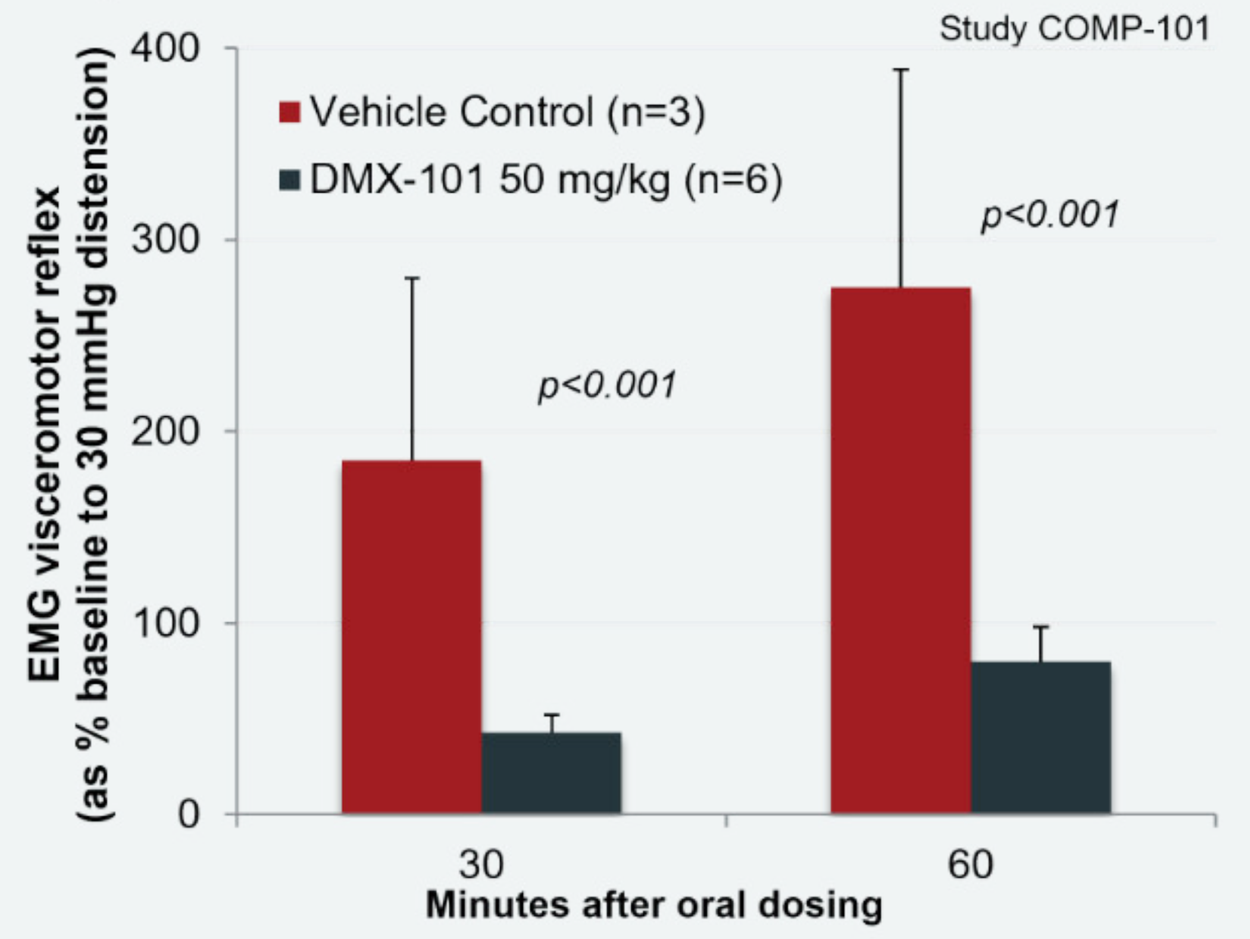
Study COMP-101
Vehicle Control (n=3)
DMX-101 50 mg/kg (n=6)
p<0.001
p<0.001
EMG visceromotor reflex (as % baseline to 30 mmHg distension)
Minutes after oral dosing

Paradigm: **Acetic acid hypersensitized hyperalgesic colorectal tissue in neonatal mouse pups**
Treatments: Single dose, oral gavage DMX-101 50 mg/kg and placebo
Assessment: Visceral myographic response (VMR) of abdominal muscles following colorectal balloon distention 30 mmHg
Time: 0, 30, 60 minutes
Results: DMX-101 controlled pain due to hypersensitivity and hyperalgesia significantly better than placebo

Study 2: Hyperalgesic Effect of DMX-101 vs. Eluxadoline and Placebo

Fig. 2b

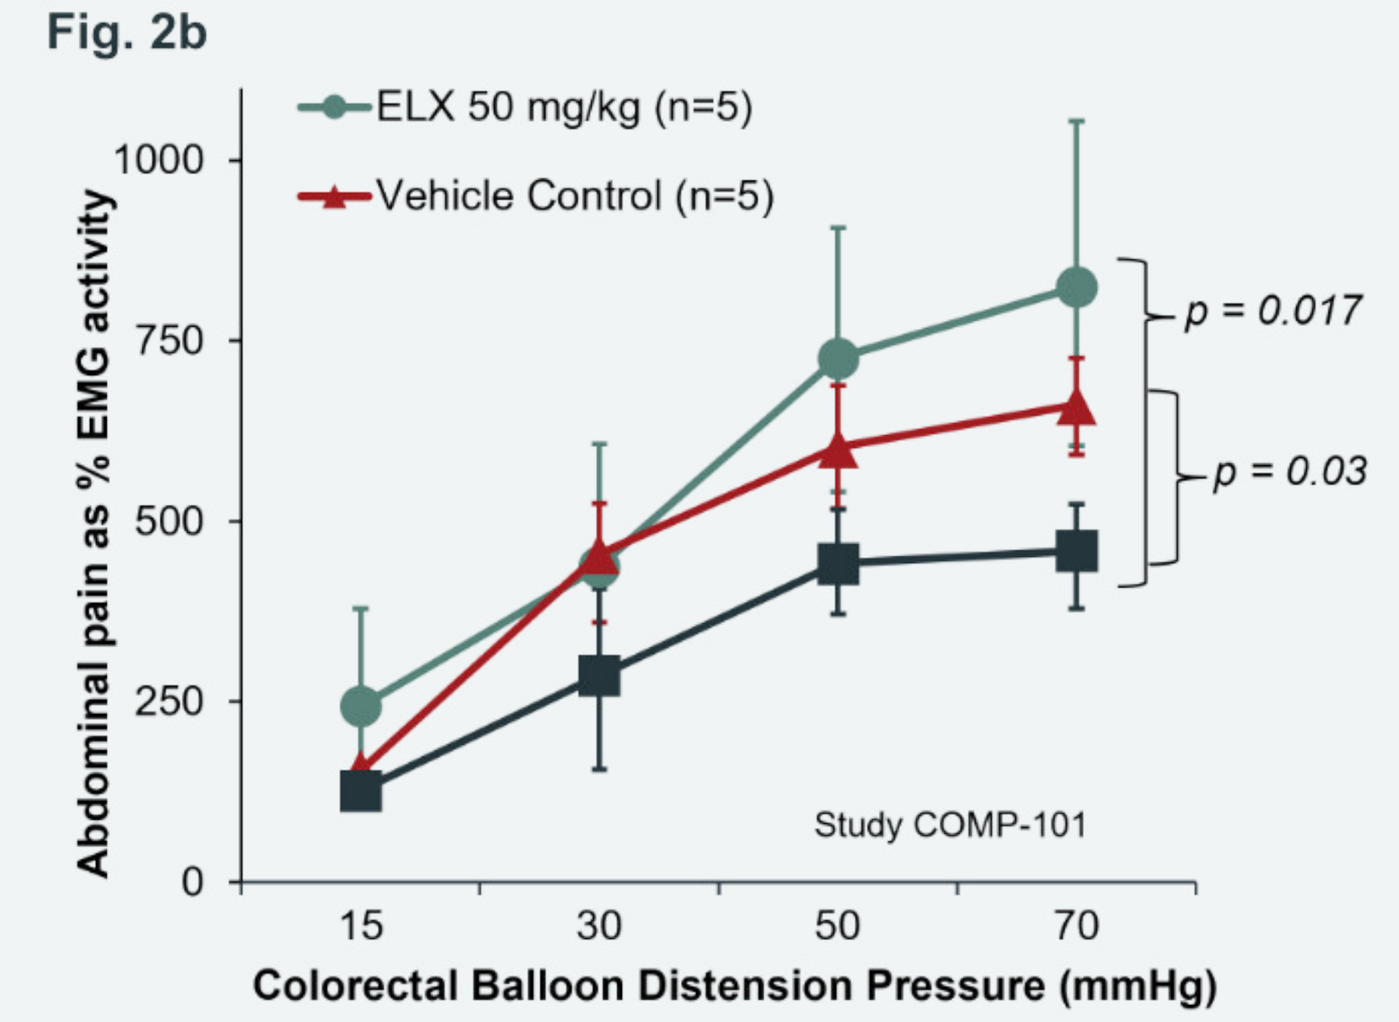
ELX 50 mg/kg (n=5)
Vehicle Control (n=5)
p = 0.017
p = 0.03
Study COMP-101
Abdominal pain as % EMG activity
Colorectal Balloon Distension Pressure (mmHg)

Paradigm: **Acetic acid hypersensitized hyperalgesic colorectal tissue in neonatal mouse pups**
Treatments: Single dose, oral gavage DMX-101 50 mg/kg, eluxadoline 50 mg/kg, and placebo
Assessment: Visceral myographic response (VMR) of abdominal muscles following colorectal balloon distention 15-70 mmHg
Time: Between 30 and 60 minutes
Results: DMX-101 controlled pain due to hypersensitivity and hyperalgesia significantly better than placebo and eluxadoline

Projected Clinical Benefit: Analgesic/antihyperalgesic agent

Paradigm: **Spinal nerve ligation at L5 and L6 in rats (Chung)**
Treatments: Single doses of subcutaneous DMX-101 10-200 mg/kg, oral gabapentin 100 mg/kg, and vehicle only
Assessment: Withdrawal response to von Frey filament pressure to the left paw
Time: Baseline, 1 hour, 3 hours
Results: DMX-101 100 mg/kg and 200 mg/kg elicited statistically significant reduction in hyperalgesia at 3 hours. These effects were comparable to that seen with 100 mg/kg oral gabapentin (threshold efficacy plasma concentration ~700 ng/mL).

Fig. 3

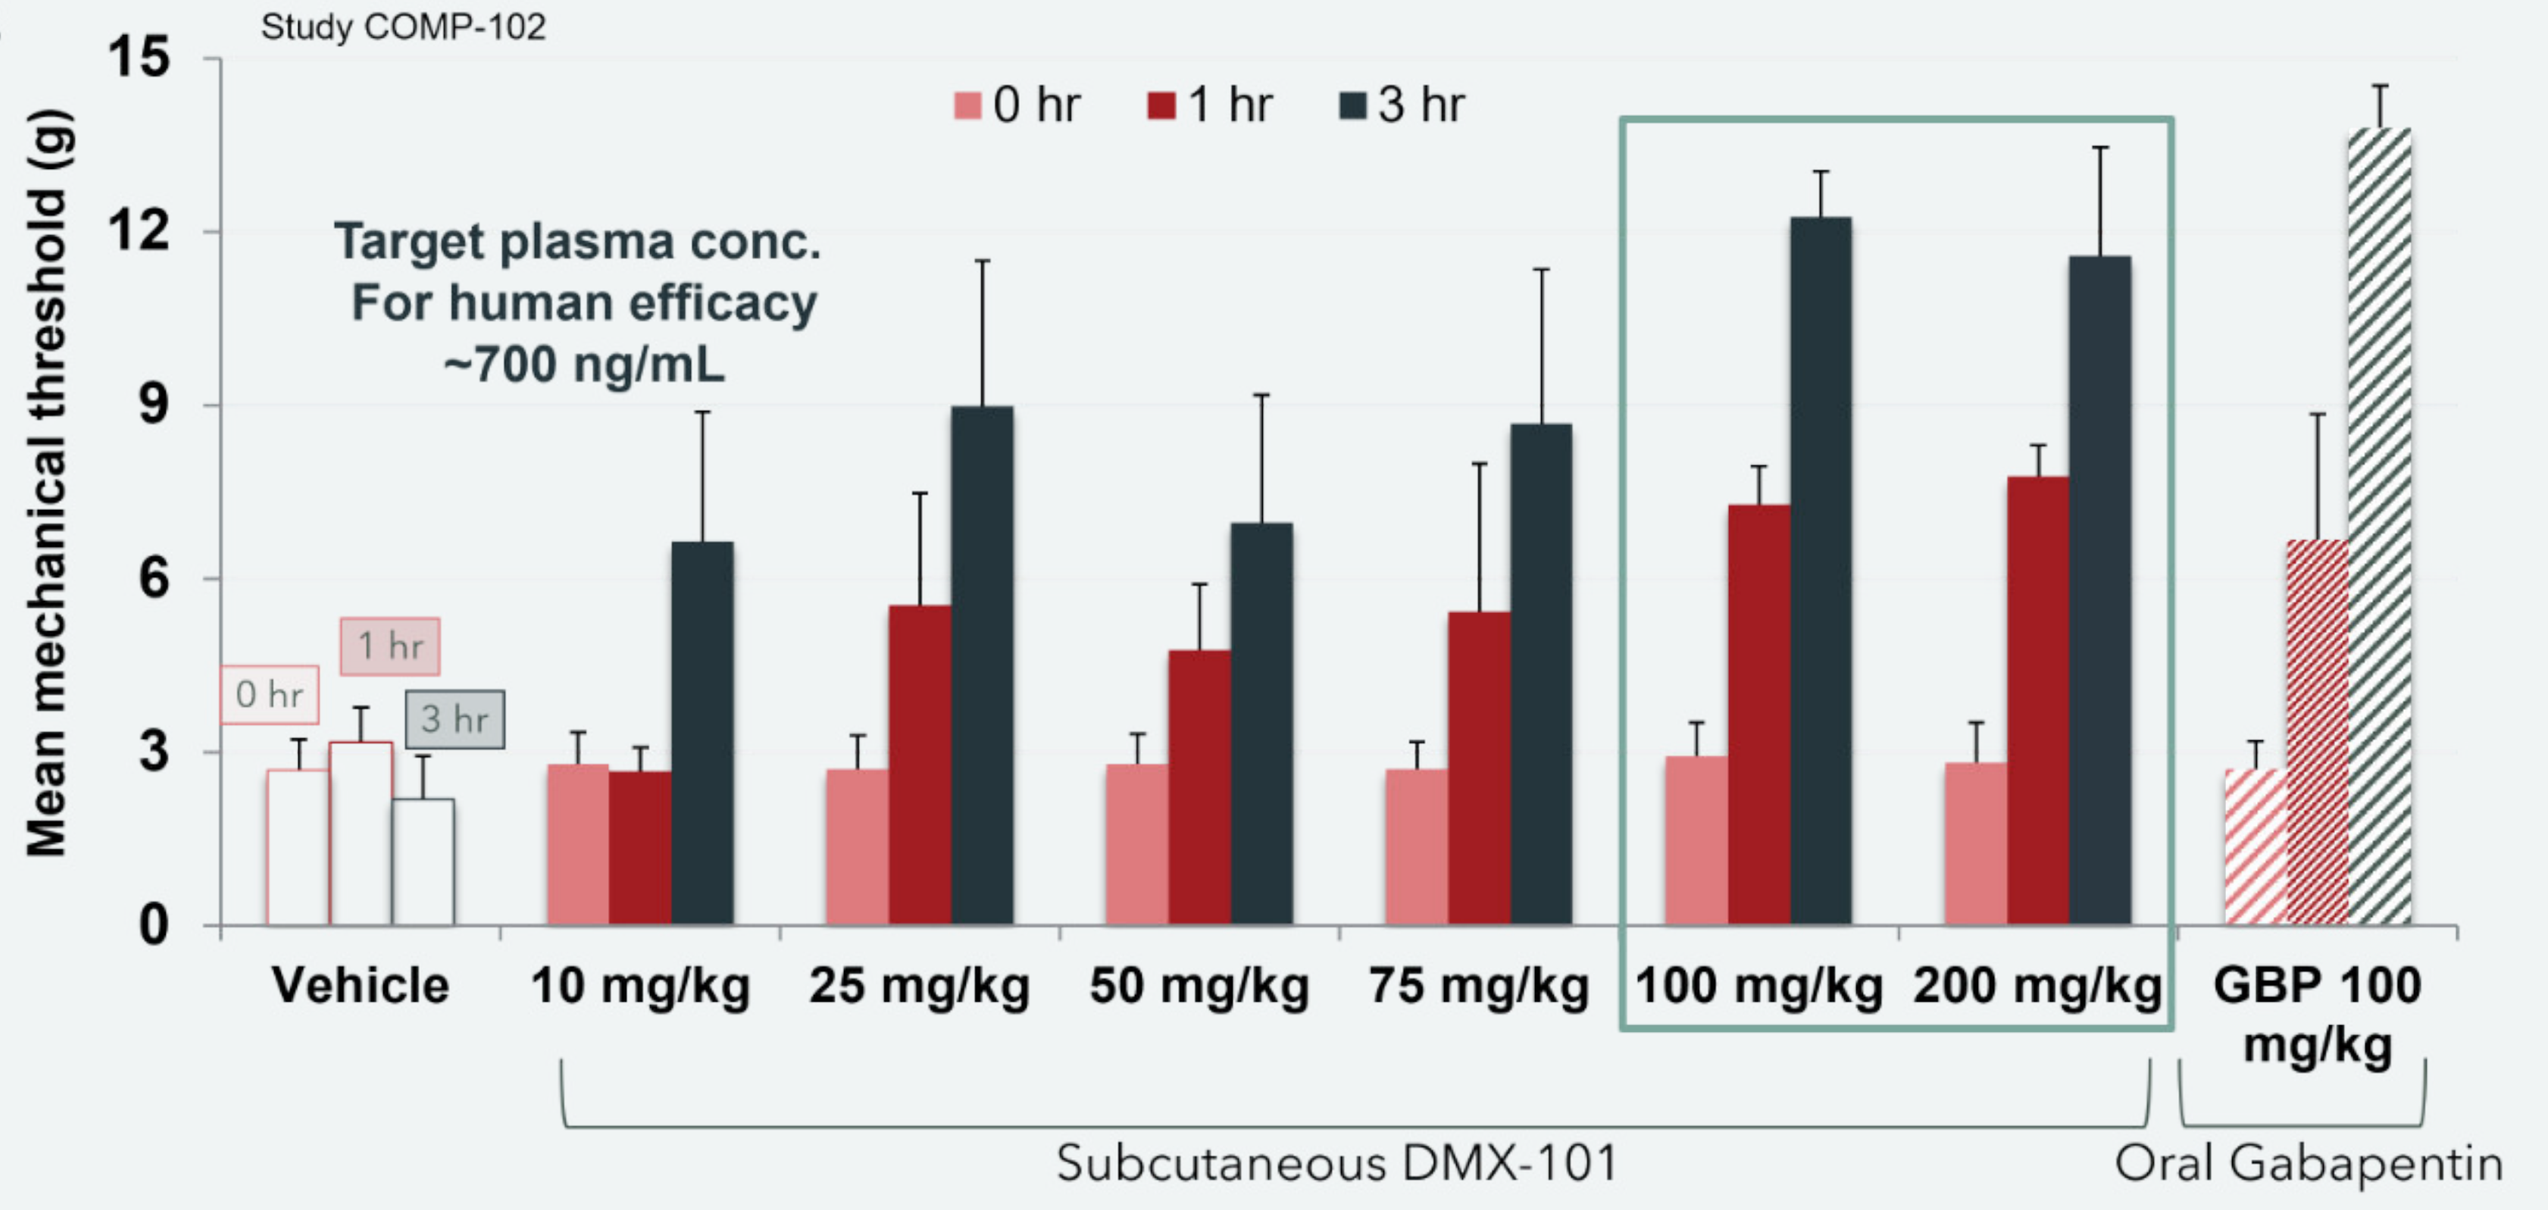
Study COMP-102
0 hr
1 hr
3 hr
Target plasma conc.
For human efficacy
~700 ng/mL
Mean mechanical threshold (g)
Vehicle
10 mg/kg
25 mg/kg
50 mg/kg
75 mg/kg
100 mg/kg
200 mg/kg
GBP 100 mg/kg
Subcutaneous DMX-101
Oral Gabapentin

Projected Clinical Benefit: Relief from neuropathic pain

Paradigm: **Sphincter of Oddi (SO) constriction model in guinea pigs**
Treatments: Single doses oral DMX-101 5 mg/kg, eluxadoline 5 mg/kg
Assessment: SO muscle tone measured by manometry
Time: Up to 25 minutes post-dose
Results: Partial mu agonist/full kappa antagonist DMX-101 decreased SO pressure relative to baseline while full mu agonist eluxadoline increased SO pressure.

Fig. 4a

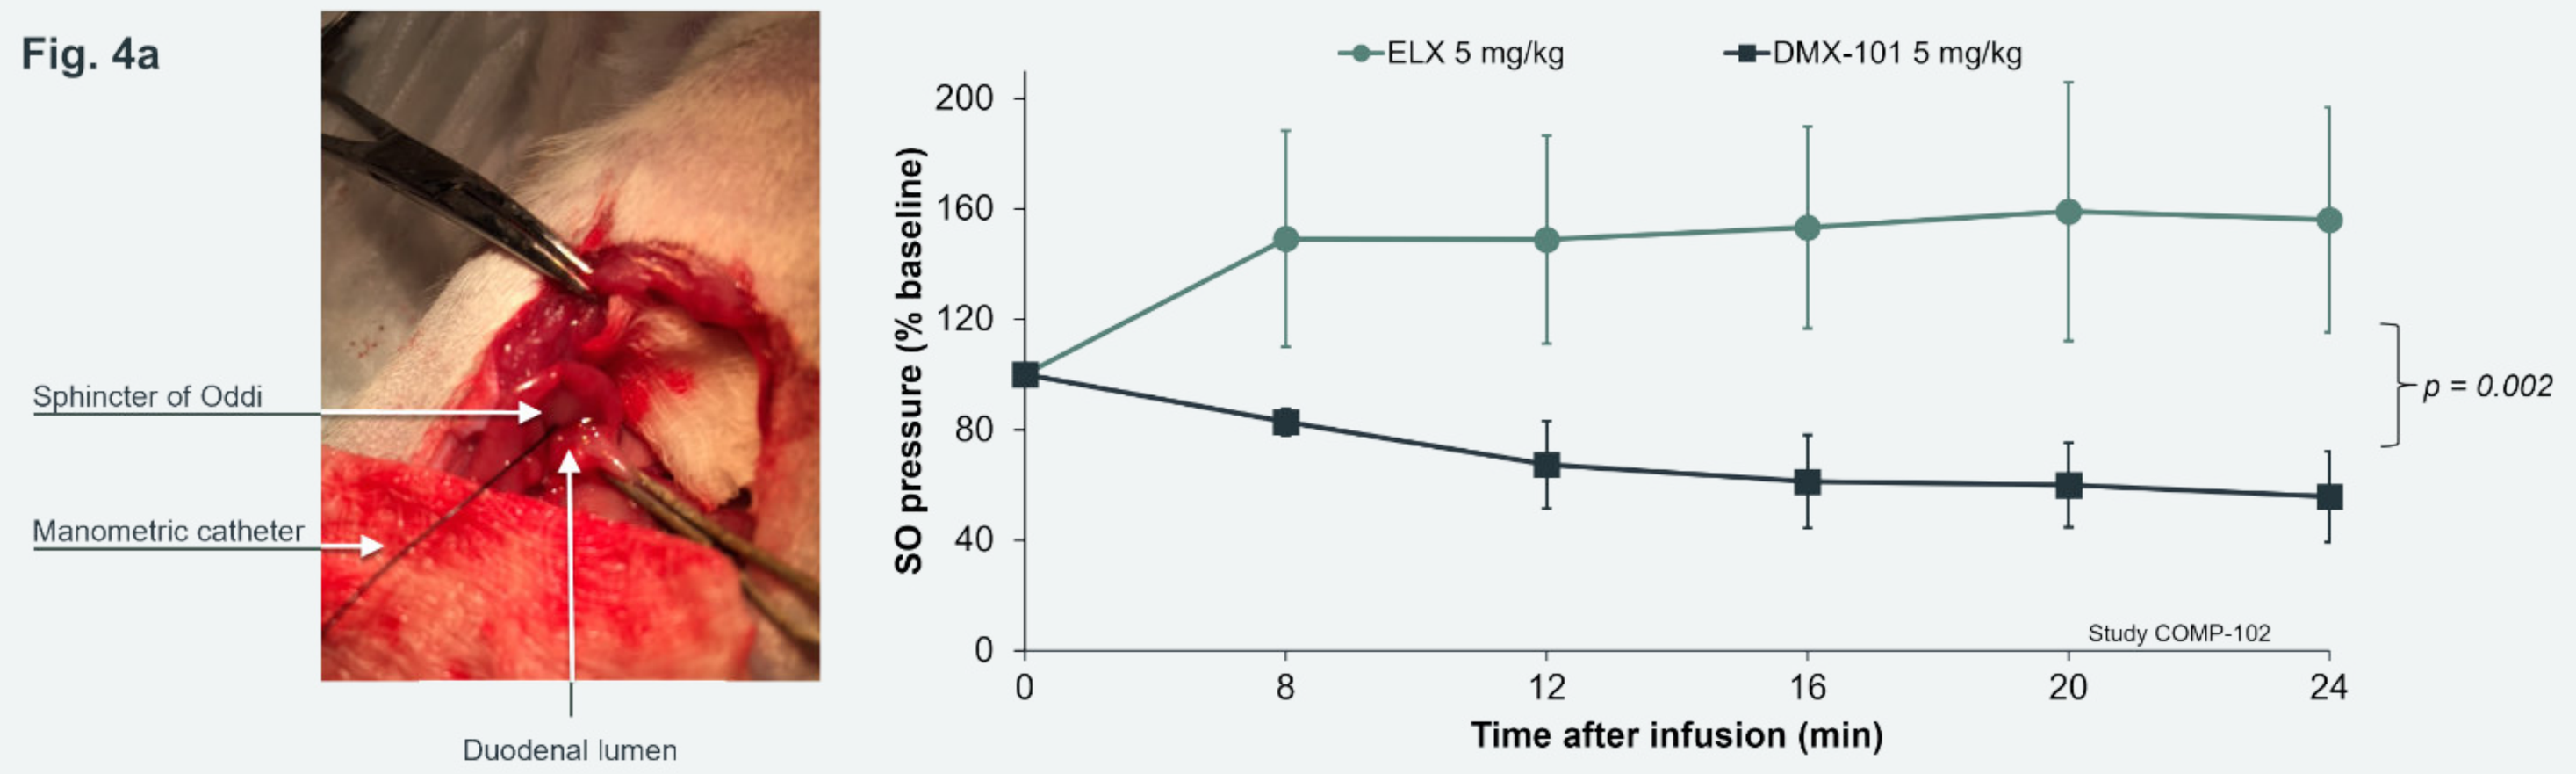
Sphincter of Oddi
Manometric catheter
Duodenal lumen
ELX 5 mg/kg
DMX-101 5 mg/kg
p = 0.002
SO pressure (% baseline)
Time after infusion (min)
Study COMP-102

Projected Clinical Benefit: No risk of pancreatitis

Paradigm: **Single ascending and multiple dose safety and pharmacokinetics studies in healthy adult volunteers**
Treatments: DMX-101 5 to 800 mg and matching placebo in single dose
DMX-101 5 to 400 mg and matching placebo in multiple dose
Assessment: Plasma levels and safety assessments including pupillary, liver function studies, and lipase and amylase levels
Time: UP to 72 hours (single dose)
Last dose (multiple dose)
Results: DMX-101 is rapidly absorbed with biomodal distribution (Figures 5a and 5b). There was no metabolism to buprenorphine. Tmax 7-8 hours; T1/2 >24 hours. At all doses, DMX-101 was safe and well tolerated. **No pupillary constriction.**

Fig. 5a

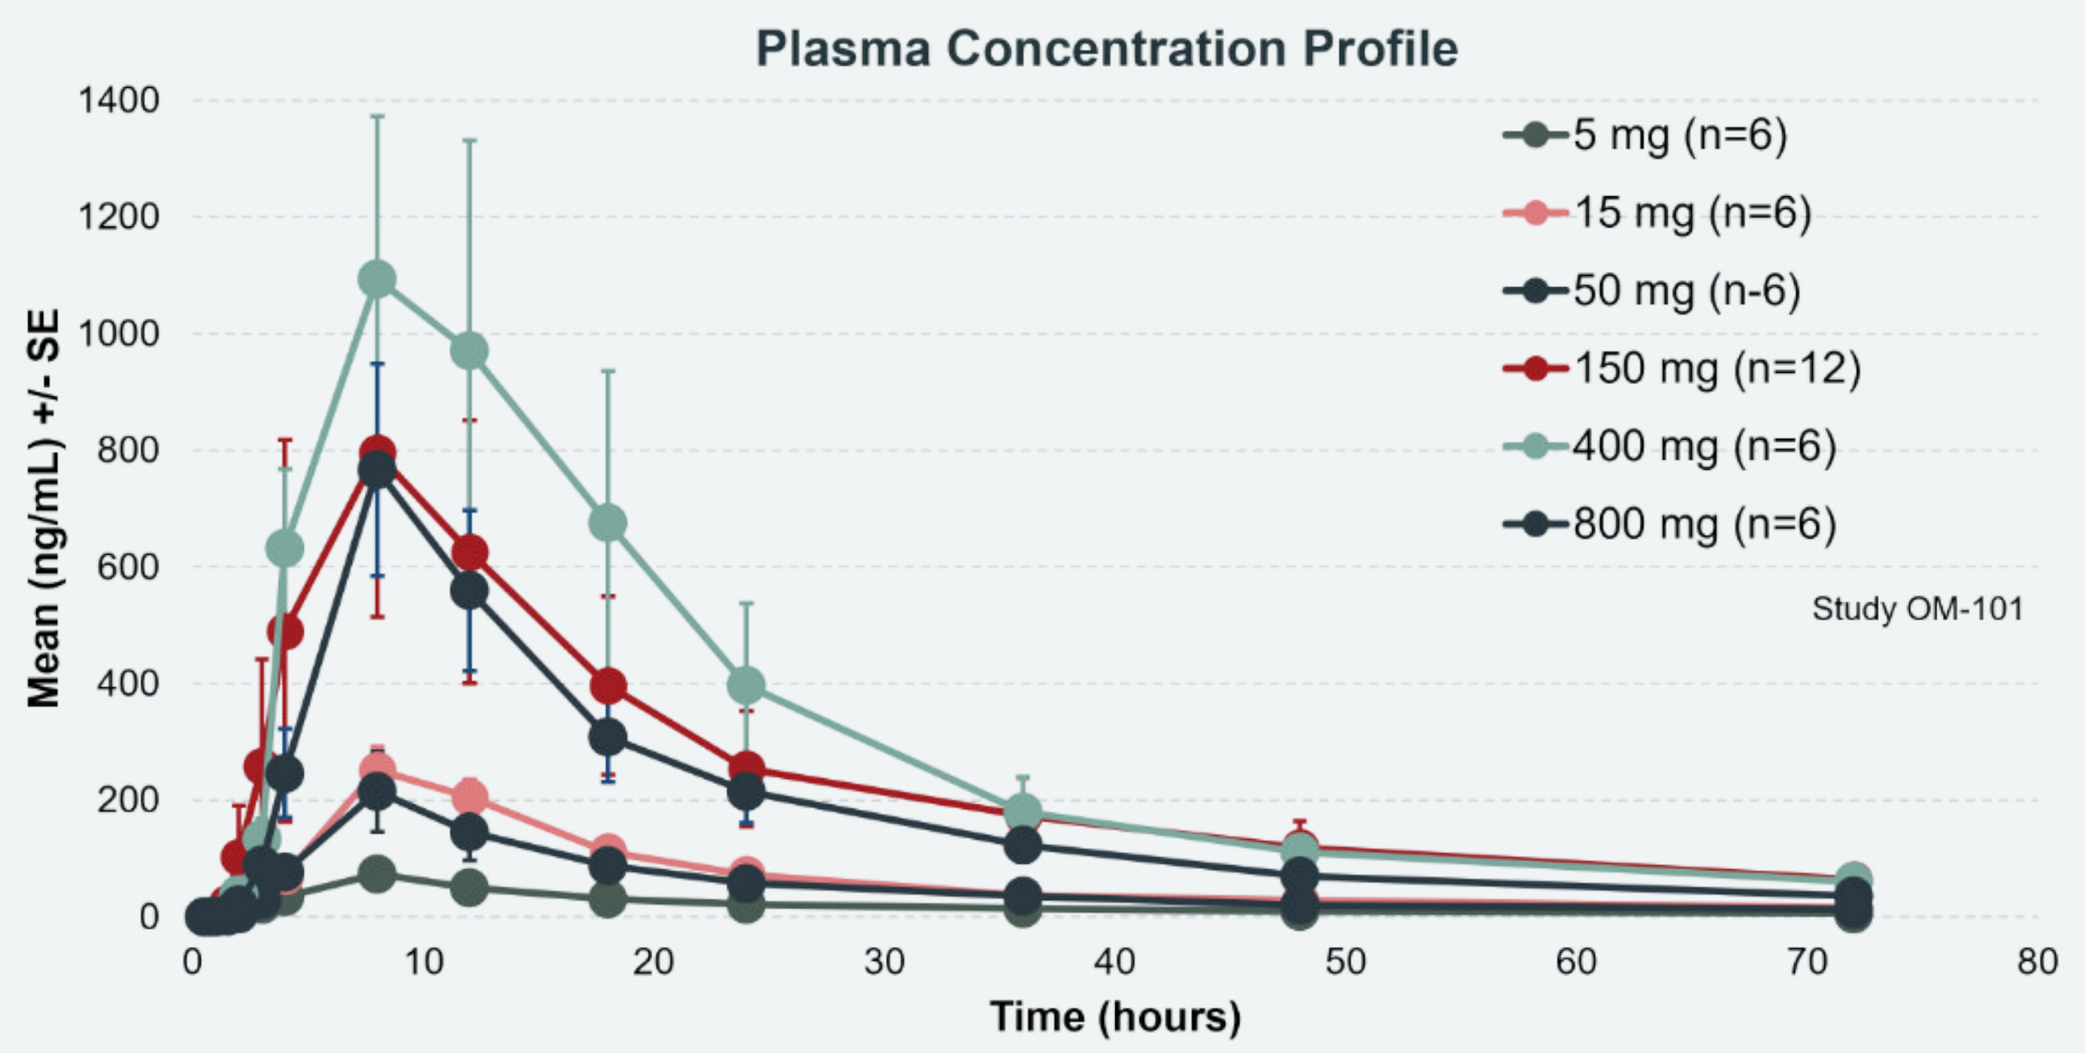
Plasma Concentration Profile
5 mg (n=6)
15 mg (n=6)
50 mg (n-6)
150 mg (n=12)
400 mg (n=6)
800 mg (n=6)
Study OM-101
Mean (ng/mL) +/- SE
Time (hours)

Fig. 5b

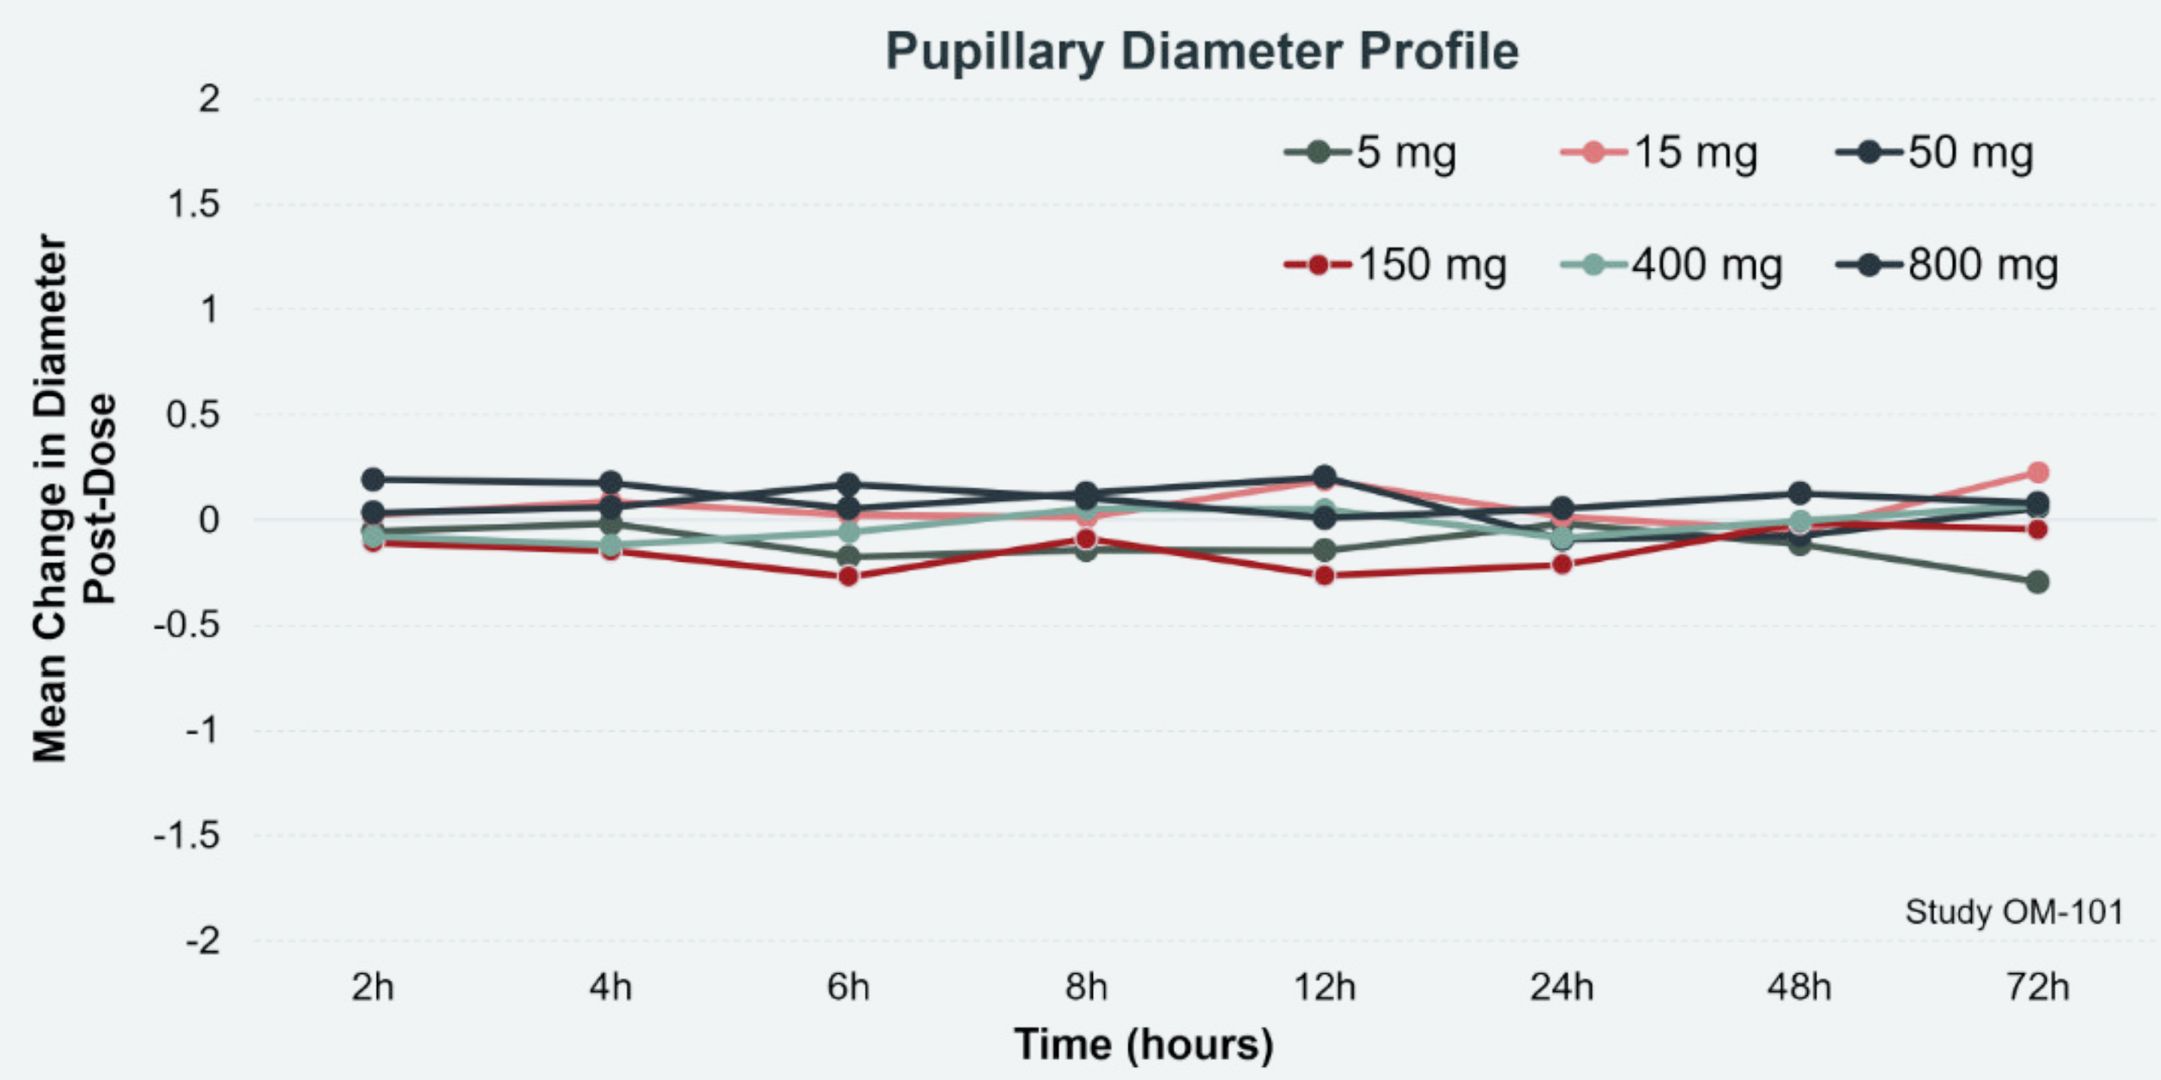
Pupillary Diameter Profile
5 mg
15 mg
50 mg
150 mg
400 mg
800 mg
Mean Change in Diameter Post-Dose
Study OM-101
Time (hours)

Absorption profile of up to 50 mg appears to be dose dependent, whereas 100 mg and above, there is saturation and a plateau in plasma levels.

Fig. 5c

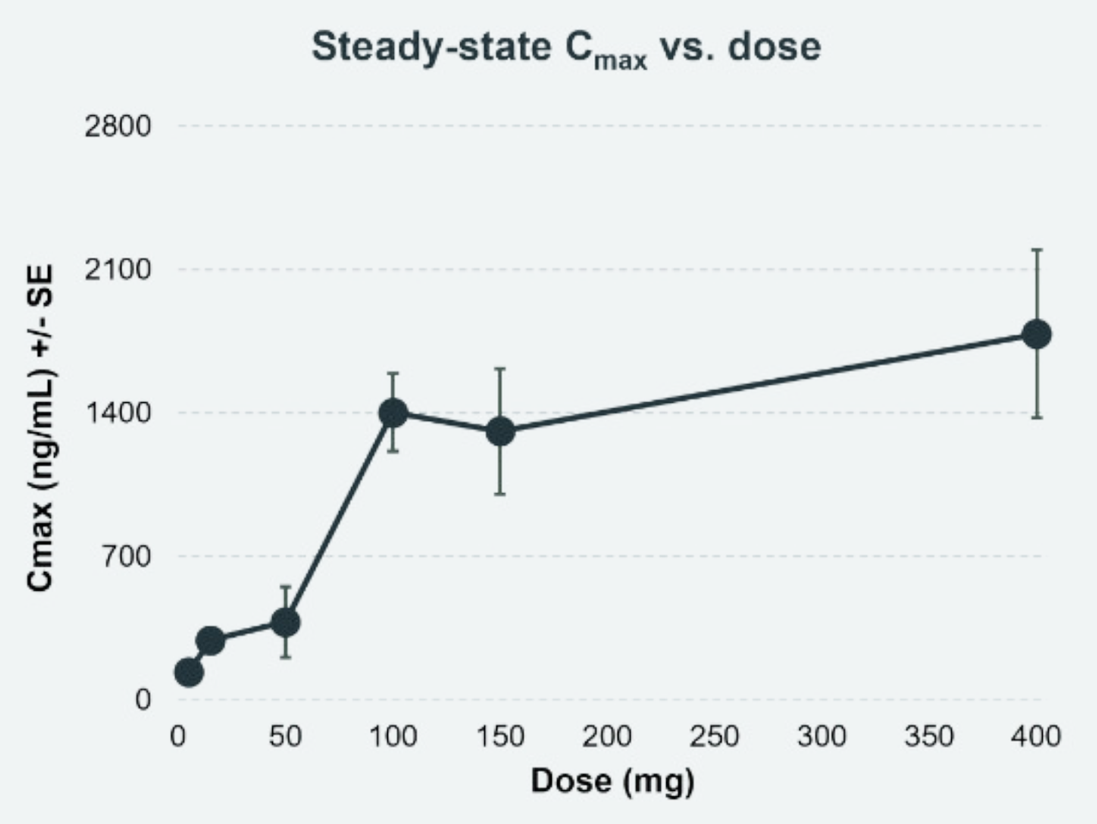
Steady-state Cmax vs. dose
Cmax (ng/mL) +/- SE
Dose (mg)

Fig. 5d

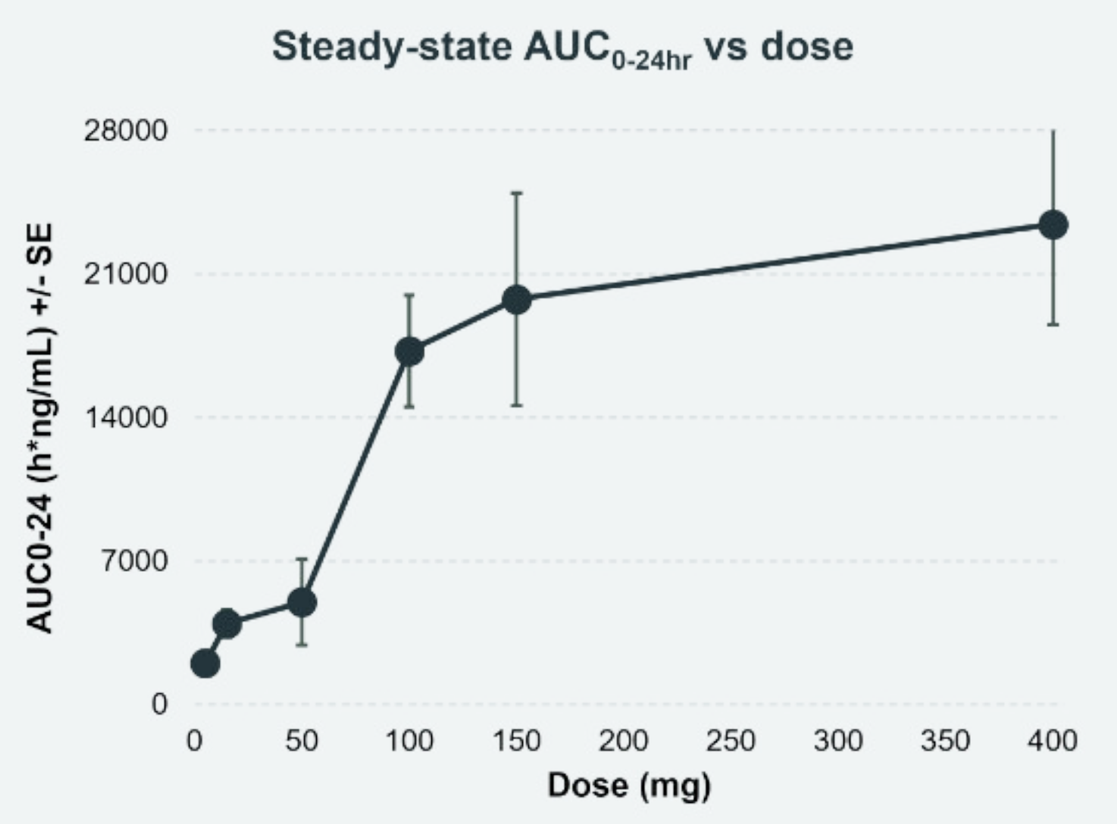
Steady-state AUC0-24hr vs dose
AUC0-24 (h*ng/mL) +/- SE
Dose (mg)

Projected Clinical Benefit:
Orally available, peripherally acting analgesic with no CNS effects

Paradigm: **Improvement in abdominal pain and stool consistency in IBS-D patients**
Treatments: Daily single dose for 12 weeks DMX-101 50 mg and 100 mg and matching placebo
Assessment: Patient impressions of clinical efficacy, quality of life, and safety assessments including pupillary constriction, liver function studies, and lipase and amylase levels
Time: Daily diary assessments (Bristol Stool scale for change in stool consistency and VAS scale change for change in abdominal pain vs. baseline)
Results: In compliant patients (target Phase 3 patient population), composite response in DMX-101 100 mg cohort was significantly better than placebo. Robust evidence of target (peripheral opioid receptor) engagement. There were no serious adverse events, including opioid-like CNS effects

Fig. 6

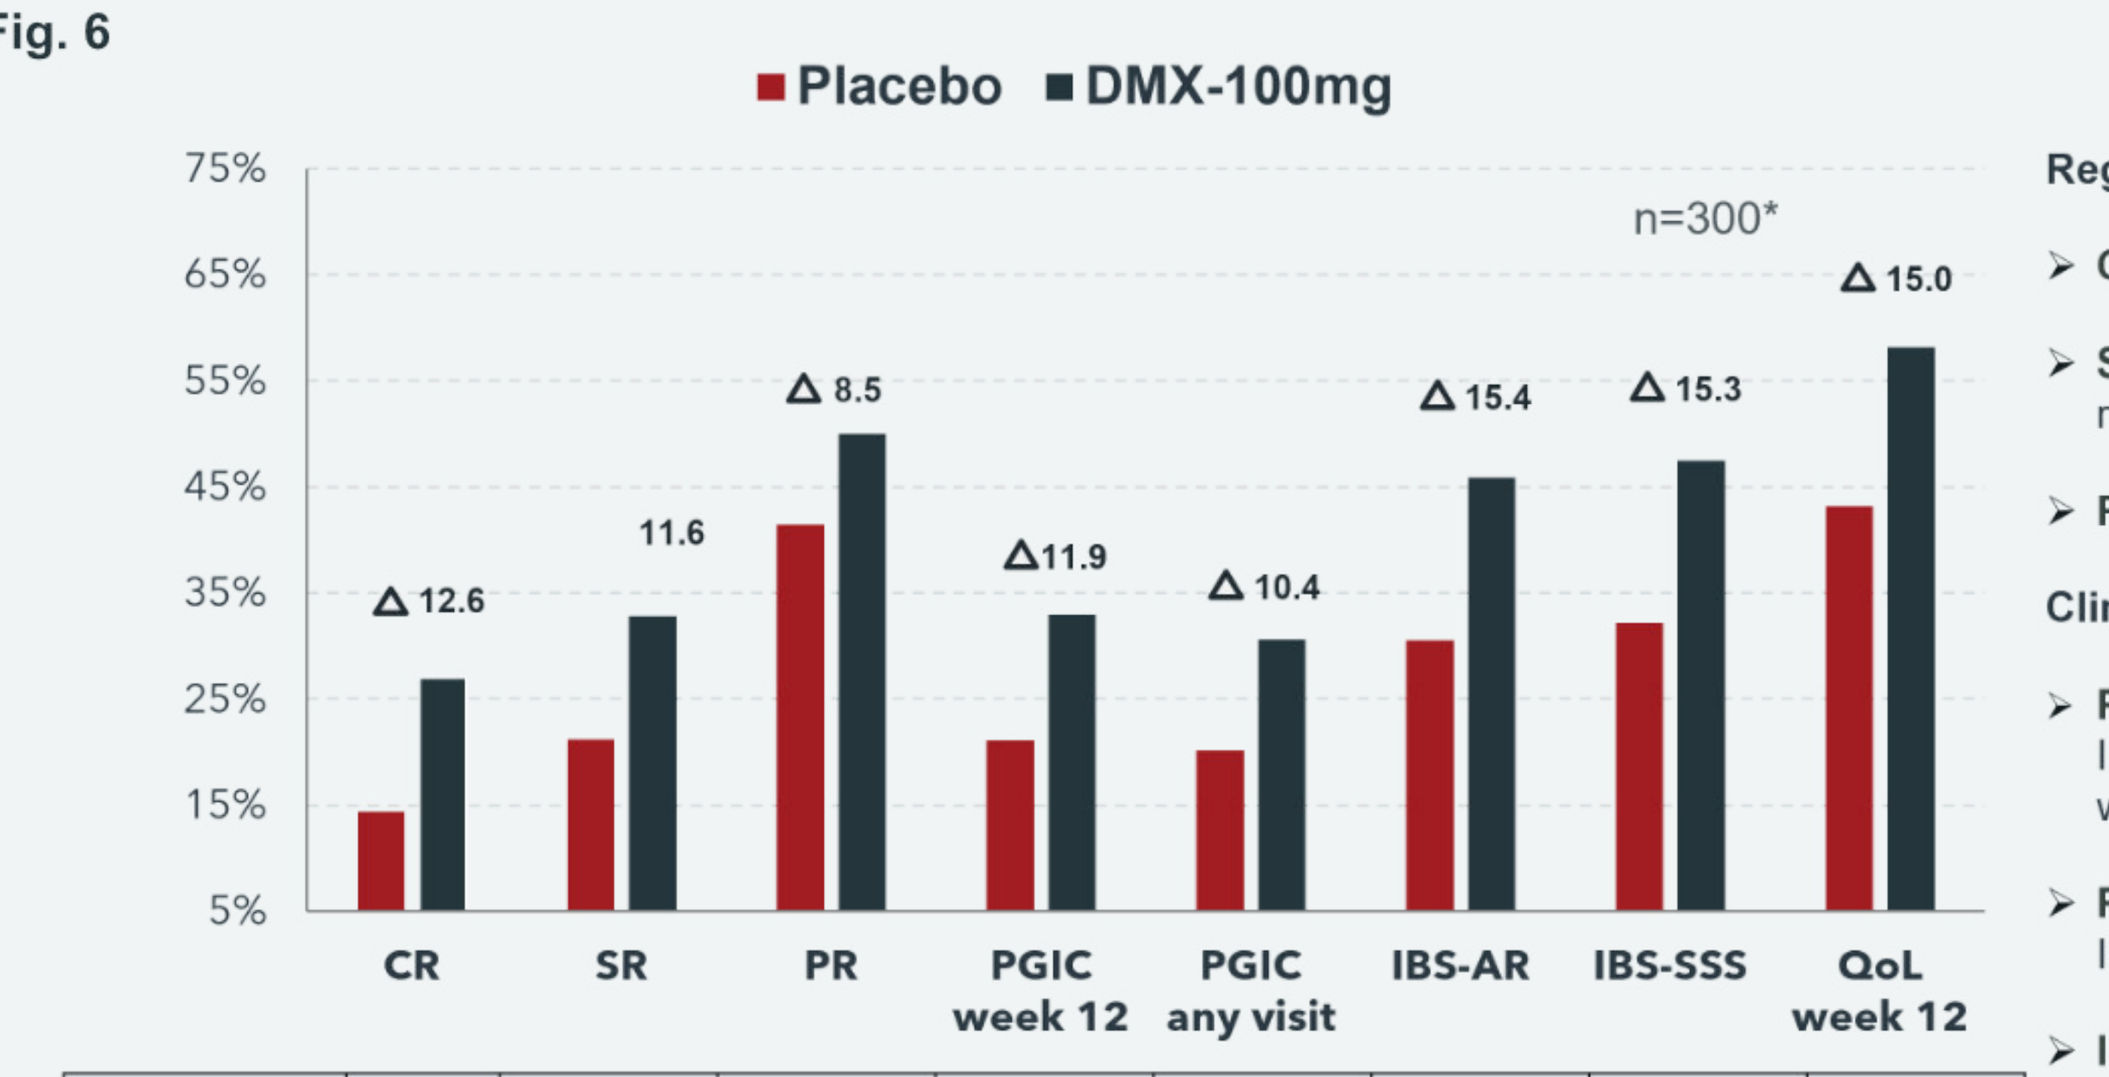
Placebo
DMX-100mg
n=300*
Δ 12.6
11.6
Δ 8.5
Δ11.9
Δ 10.4
Δ 15.4
Δ 15.3
Δ 15.0
CR
SR
PR
PGIC week 12
PGIC any visit
IBS-AR
IBS-SSS
QoL week 12

Arm	CR	SR	PR	PGIC week 12	PGIC any visit	IBS-AR	IBS-SSS	QoL week 12
100mg (n=122)	**27.0%**	**32.8%**	**50.0%**	**33.0%**	**30.6%**	**45.9%**	**47.5%**	**58.2%**
Placebo (n=118)	14.4%	21.2%	41.5%	21.1%	20.2%	30.5%	32.2%	43.2%
Delta	12.6	11.6	8.5	11.9	10.4	15.4	15.3	15.0
P-value	**0.008**	**0.022**	**0.093**	**0.031**	**0.040**	**0.007**	**0.008**	**0.010**

*Analysis excludes COVID-19 affected patients and evaluates those reporting excessive use of rescue medication as non-responders

Study OM-201

Legend

Regulatory Endpoints:

- **CR:** Composite responder rate
- **SR:** Stool consistency responder rate
- **PR:** Pain responder rate

Clinical Endpoints:

- **PGIC-week 12:** Patient Global Impression of Change (at 12 weeks)
- **PGIC-any visit:** Patient Global Impression of Change (at any visit)
- **IBS-AR:** IBS-Adequate Relief
- **IBS-SSS:** IBS-Symptom Severity Score
- **QoL week 12:** Improvement in IBS Quality of Life at week 12 / early termination visit

Evidence of engagement with peripheral opioid receptors

Conclusion

DMX-101, a new chemical entity, demonstrated safety and robust engagement with peripheral opioid receptors in preclinical and clinical studies without the unwanted CNS side effects or acute pancreatitis risk of traditional opioids.

Potential applications include:

- Chronic pain
- Gastrointestinal disorders

Societal Impact: Potential to decrease reliance on addictive opioids and prevent future cases of opioid abuse and dependence.

Bibliography

1. Iwaszkiewicz, K. S., Schneider, J. J. & Hua, S. Targeting peripheral opioid receptors to promote analgesic and anti-inflammatory actions. *Front Pharmacol* 4, 132 (2013). doi:10.3389/fphar.2013.00132. PMCID: PMC3807052.
2. DeHaven-Hudkins, D. L. & Dolle, R. E. Peripherally restricted opioid agonists as novel analgesic agents. *Curr Pharm Des* 10, 743-757 (2004). doi:10.2174/1381612043453036.
3. Vadivelu, N., Mitra, S. & Hines, R. L. Peripheral opioid receptor agonists for analgesia: a comprehensive review. *J Opioid Manag* 7, 55-68 (2011). doi:10.5055/jom.2011.0049.
4. Stein, C. *et al.* Peripheral mechanisms of pain and analgesia. *Brain Res Rev* 60, 90-113 (2009). doi:10.1016/j.brainresrev.2008.12.017. PMCID:PMC2730351.